Exposure-Response Analyses of Sacituzumab Govitecan Efficacy and Safety in Patients With Metastatic Breast Cancer

Poster # PO1-04-06

Indrajeet Singh,[1*] Abhishek G. Sathe,[1] Paul M. Diderichsen,[2] Han Witjes,[2] Arne Van Schanke,[2] John Maringwa,[2*] Wendy Verret,[3] Sandhya Girish,[1] Ahmed A. Othman[1]

[1]Clinical Pharmacology, Gilead Sciences, Inc., Foster City, CA, USA; [2]Integrated Drug Development Consulting, Certara USA, Inc., Princeton, NJ, USA; [3]Clinical Research, Gilead Sciences, Inc., Foster City, CA, USA

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from SABCS® and the author of this poster

San Antonio Breast Cancer Symposium® - December 5-9, 2023

Key Findings

- **SG efficacy and safety end points of clinical interest were correlated with SG serum exposures in patients with mBC**
- **The average SG serum exposures over the treatment duration until the end point of interest were consistently identified as the most significant exposure metric correlated with CBR, CR, and ORR in pts with HR+/HER2– mBC, and with select AEs of diarrhea, neutropenia, nausea, vomiting, and hypersensitivity in pts with mBC (mTNBC or HR+/HER2– mBC)**
- **Within the evaluated exposure range, higher values of total antibody average serum exposures were associated with significantly longer PFS and OS**

Conclusions

The clinically desirable efficacy and manageable safety profile of the 10 mg/kg SG dose on days 1 and 8 of the 21-day cycle support the appropriateness of this regimen in pts with HR+/HER2– mBC

References: **1.** TRODELVY® https://www.gilead.com/-/media/files/pdfs/medicines/oncology/trodelvy/trodelvy_pi.pdf. Accessed September 2023. **2.** Rugo HS, et al. *Lancet*. 2023;402(10411):1423-1433. **3.** Bardia A, et al. *Ann Oncol.* 2021;32:746-56. **4.** Bardia A, et al. *N Engl J Med.* 2021;384:1529-1541. **5.** Singh I, et al. *J Clin Oncol.* 2022;40(suppl 16):1076. **6.** Sathe AG, et al. *Clin Pharm Ther.* 2023;113 (suppl S1):S70.

Acknowledgments: We thank the investigators, patients and their caregivers for their participation and commitment to clinical research. This study is sponsored by Gilead Sciences, Inc. Editorial support was provided by Sam Phillips, PhD, of Parexel and funded by Gilead Sciences, Inc.

*At the time of this work.

Disclosures: Indrajeet Singh was an employee of Gilead Sciences, Inc. at the time the work was undertaken. Abhishek Sathe declares employment, travel expenses, and stocks from Gilead Sciences, Inc.

Correspondence: abhishek.sathe@gilead.com

NOTE: HR+/HER2– (IHC 0, 1+ or 2+/ISH–), hormone receptor-positive/human epidermal growth factor receptor 2-negative (immunohistochemistry 0, 1+ or 2+/in situ hybridization-negative).

Introduction

- Sacituzumab govitecan (SG) is a Trop-2–directed antibody-drug conjugate (ADC); it contains a hydrolyzable CL2A linker that facilitates rapid internalization of potent SN-38 (the active metabolite of the topoisomerase inhibitor irinotecan) to tumor cells, and into the surrounding tumor microenvironment via the bystander effect[1]
- SG is approved for unresectable, locally advanced or metastatic triple-negative breast cancer (mTNBC) following ≥ 2 prior systemic therapies (≥ 1 in the metastatic setting) and for HR+/HER2– (IHC 0, IHC 1+, or IHC 2+/ISH–) metastatic breast cancer (mBC) following endocrine-based therapy and ≥ 2 additional systemic therapies in the metastatic setting[1]

Objective

- These analyses characterized the relationship between the exposure of SG (ADC), free SN-38, and total antibody (tAB) and the efficacy outcomes in 260 patients (pts) with HR+/HER2– mBC from TROPiCS-02 (NCT03901339) and safety outcomes in all 569 pts with mBC (mTNBC or HR+/HER2– mBC) from IMMU-132-01 (NCT01631552), ASCENT (NCT02574455), and TROPiCS-02[2-4]

Methods

- The designs of the 3 clinical studies used for these analyses are presented in **Figure 1**
- Exposure-response models were developed to characterize the relationship between exposure and efficacy or safety using previously described methods.[5] Four exposure metrics for each analyte (SG, free SN-38, and tAB) based on the previously described population pharmacokinetic models,[6] including average concentrations (CAVG) between the first dose and time of event, maximum concentration and area under the concentration-time curve during first treatment cycle, and cumulative exposure until the time of event, were assessed
- Impact of exposure on efficacy end points was evaluated using logistic regression for clinical benefit rate (CBR), complete response (CR) and objective response rate (ORR), and Cox proportional hazard models for progression-free survival (PFS) and overall survival (OS). Impact of exposure on safety end points was evaluated using categorical logistic regression with nonproportional odds for adverse events (AEs; diarrhea, hypersensitivity, nausea, neutropenia/febrile neutropenia, and vomiting) or Cox proportional hazard models (time to first dose delay or reduction). The effects of clinically relevant covariates (eg, Trop-2 expression, *UGT1A1* genotype) were assessed in a stepwise covariate model
- Data for the most significant metric per outcome are presented

Figure 1. Study designs

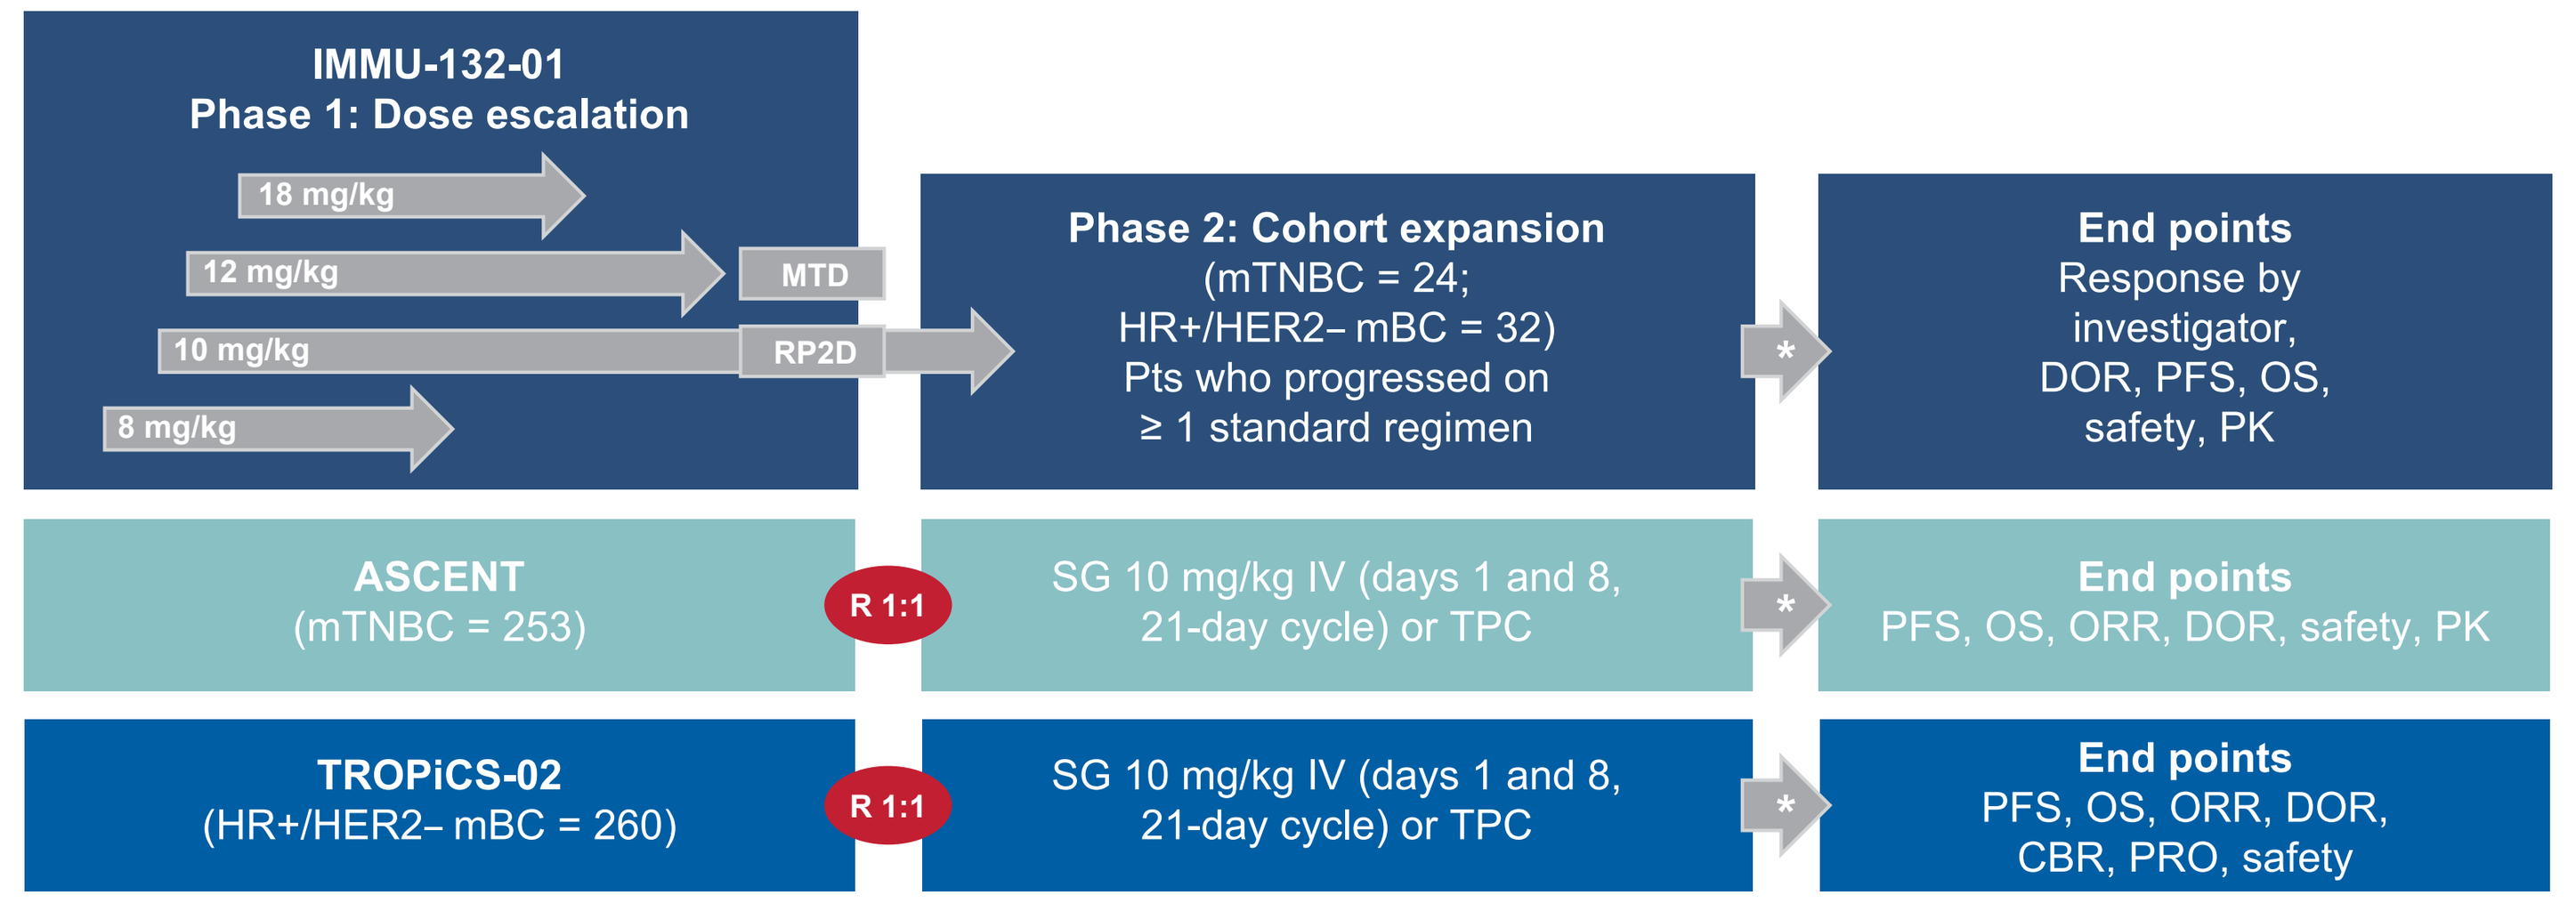
IMMU-132-01
Phase 1: Dose escalation
18 mg/kg
12 mg/kg
MTD
10 mg/kg
RP2D
8 mg/kg
Phase 2: Cohort expansion
(mTNBC = 24;
HR+/HER2– mBC = 32)
Pts who progressed on
≥ 1 standard regimen
*
End points
Response by
investigator,
DOR, PFS, OS,
safety, PK
ASCENT
(mTNBC = 253)
R 1:1
SG 10 mg/kg IV (days 1 and 8,
21-day cycle) or TPC
End points
PFS, OS, ORR, DOR, safety, PK
TROPiCS-02
(HR+/HER2– mBC = 260)
R 1:1
SG 10 mg/kg IV (days 1 and 8,
21-day cycle) or TPC
End points
PFS, OS, ORR, DOR,
CBR, PRO, safety

CBR, clinical benefit rate; DOR, duration of response; IV, intravenous; mBC, metastatic breast cancer; MTD, maximum tolerated dose; mTNBC, metastatic triple-negative breast cancer; ORR, objective response rate; OS, overall survival; PFS, progression-free survival; PK, pharmacokinetics; PRO, patient-reported outcomes; Pts, patients; R, randomized; RP2D, recommended phase 2 dose; SG, sacituzumab govitecan; TPC, treatment of physicians' choice. *Treatment continuation until progression or unacceptable toxicity.

Results

Exposure-efficacy analysis

- Baseline characteristics of pts by study are summarized in **Table 1**
- Within the evaluated exposure range, higher $CAVG_{tAB}$ values were associated with significantly longer PFS and OS **(Table 2, Table 3,** and **Figures 2C** and **2D)**. Higher baseline lactate dehydrogenase was associated with shorter OS (data not shown). No additional effects of covariates were identified in the PFS model. Higher $CAVG_{SG}$ values were associated with increased probability of CBR, CR, and ORR **(Table 2, Figure 2A)**. No additional effects of covariates were identified

Table 1. Demographics and baseline characteristics

Inclusion criteria	TROPiCS-02 (N = 260)	IMMU-132-01 (N = 56)	ASCENT (N = 253)
Age, median (range), years	58 (29-86)	53 (31-79)	54 (27-82)
Female, n (%)	259 (100)	56 (100)	251 (99)
LDH, median (range), IU/L	263 (129-2040)	274 (114-2190)	283 (107-5930)
Body weight, median (range), kg	67 (40-128)	62 (42-125)	68 (37-132)
Trop-2 expression, median (range)	125 (0-289)	NA	220 (0-300)
Prior chemotherapies,[a] median (range)	3 (0-8)	NA	3 (1-15)
ECOG PS 0, n (%)	108 (42)	24 (43)	112 (44)
ECOG PS 1, n (%)	152 (58)	32 (57)	138 (55)
ECOG PS 2, n (%)	0	0	3 (1)
UGT1A1 genotype *1/*1, n (%)[b]	98 (38)	19 (34)	110 (43)
UGT1A1 genotype *1/*28, n (%)[b]	116 (45)	21 (38)	95 (38)
UGT1A1 genotype *28/*28, n (%)[b]	24 (9)	8 (14)	34 (13)

ECOG PS, Eastern Cooperative Oncology Group performance status; LDH, lactate dehydrogenase; NA, not available.
[a]In the metastatic setting. [b]Excludes other genotypes or missing data.

Table 2. Model-predicted probabilities for efficacy end points in pts receiving SG (starting dose 10 mg/kg)

End point	Probability (95% CI)	End point (at 6 months)	Survival probability at mean $CAVG_{tAB}$
CBR	0.331 (0.285-0.381)	PFS	0.438 (0.369-0.508)
CR	0.008 (0-0.016)	OS	0.877 (0.838-0.915)
ORR	0.204 (0.162-0.250)		

$CAVG_{tAB}$, total antibody average concentration; CBR, clinical benefit rate; CI, confidence interval; CR, complete response; ORR, objective response rate; OS, overall survival; PFS, progression-free survival; pts, patients; SG, sacituzumab govitecan.
PFS based on the primary analysis. OS based on second interim analysis.

Table 3. Median survival times for PFS and OS in quartiles of $CAVG_{tAB}$

End point	Quartile	Median $CAVG_{tAB}$ (range), µg/mL	Median survival (95% CI), months
PFS	1	95 (55-116)	3.25 (2.79-4.27)
	2	135 (117-150)	2.86 (2.56-5.45)
	3	170 (150-190)	5.58 (4.17-10.3)
	4	232 (190-568)	9.00 (8.51-12.5)
OS	1	88 (48-110)	9.06 (6.67-11.6)
	2	127 (110-143)	12.0 (9.82-14.4)
	3	169 (145-187)	16.9 (13.9-22.7)
	4	226 (187-568)	26.8 (21.9-NA)

$CAVG_{tAB}$, total antibody average concentration; CI, confidence interval; NA, not available; OS, overall survival; PFS, progression-free survival.
PFS based on the primary analysis. OS based on second interim analysis.

Figure 2. Observed proportion and predicted probability[a] of ORR vs $CAVG_{SG}$ (A). Predicted probability of any grade neutropenia vs $CAVG_{SG}$ by *UGT1A1* genotype (B). Kaplan-Meier curves of PFS (C) and OS (D) stratified by $CAVG_{tAB}$ quartiles[b]

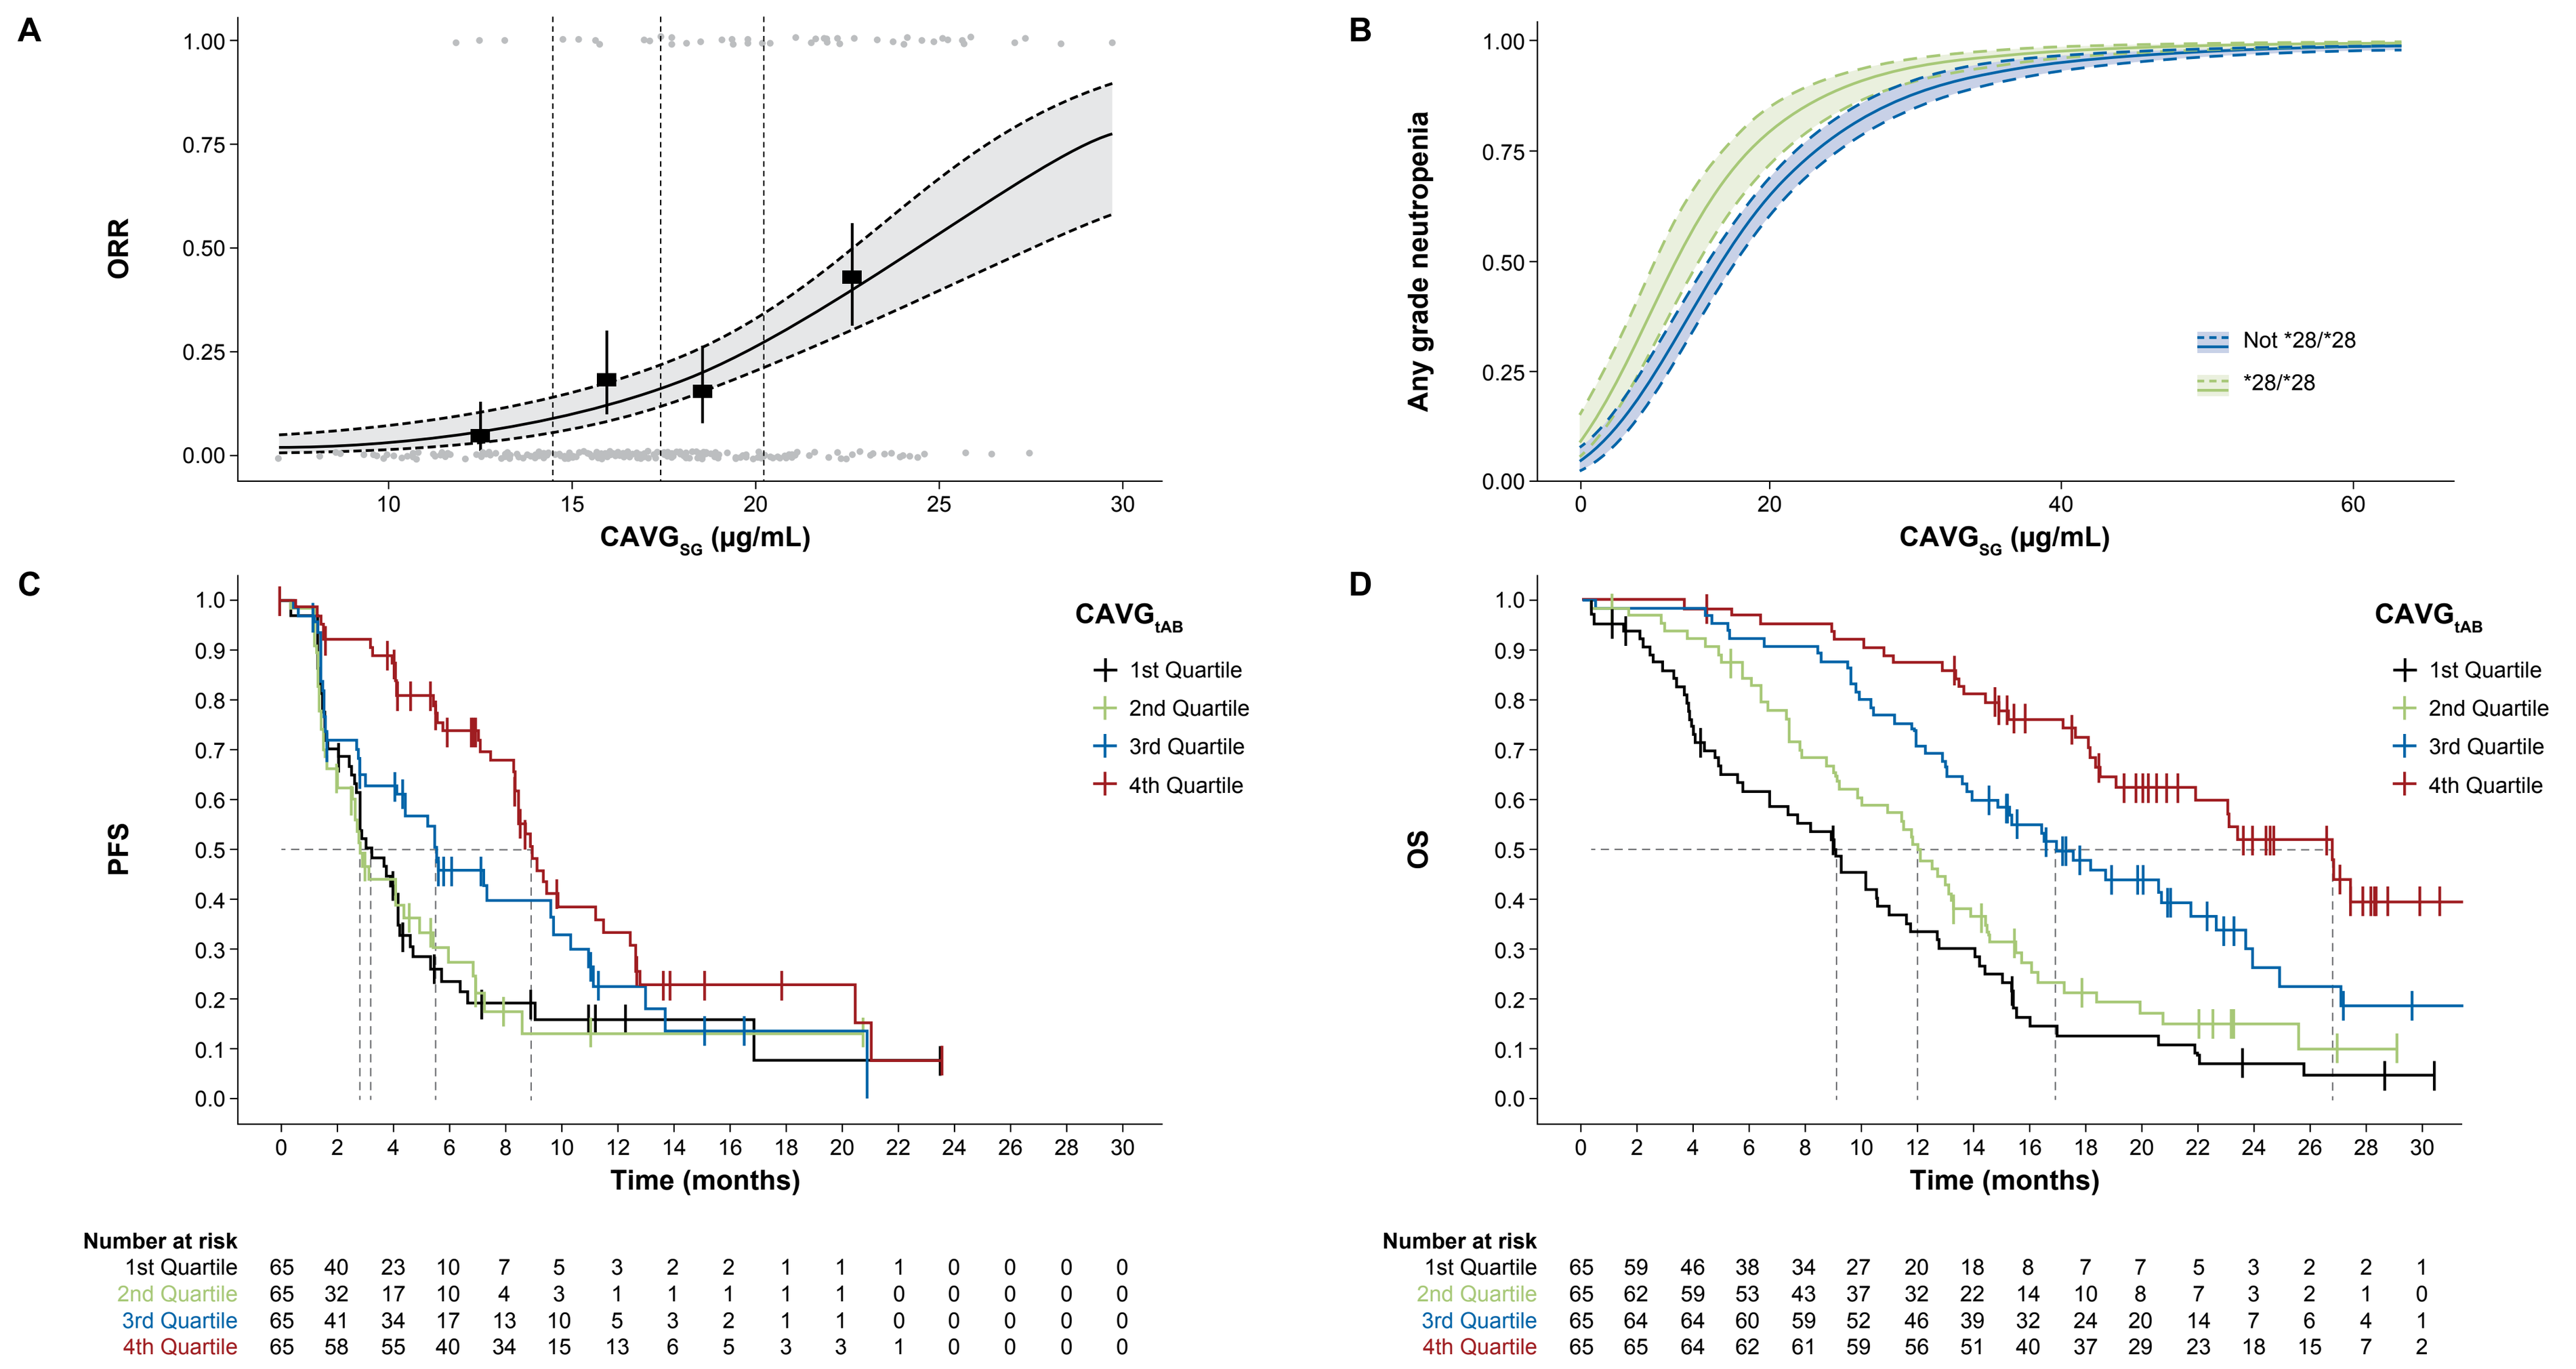
A
ORR
1.00
0.75
0.50
0.25
0.00
10
15
20
25
30
CAVGSG (µg/mL)
B
Any grade neutropenia
1.00
0.75
0.50
0.25
0.00
0
20
40
60
Not *28/*28
*28/*28
CAVGSG (µg/mL)
C
PFS
CAVGtAB
1st Quartile
2nd Quartile
3rd Quartile
4th Quartile
Time (months)
0 2 4 6 8 10 12 14 16 18 20 22 24 26 28 30
Number at risk
1st Quartile 65 40 23 10 7 5 3 2 2 1 1 1 0 0 0 0
2nd Quartile 65 32 17 10 4 3 1 1 1 1 1 0 0 0 0 0
3rd Quartile 65 41 34 17 13 10 5 3 2 1 1 0 0 0 0 0
4th Quartile 65 58 55 40 34 15 13 6 5 3 3 1 0 0 0 0
D
OS
CAVGtAB
1st Quartile
2nd Quartile
3rd Quartile
4th Quartile
Time (months)
0 2 4 6 8 10 12 14 16 18 20 22 24 26 28 30
Number at risk
1st Quartile 65 59 46 38 34 27 20 18 8 7 7 5 3 2 2 1
2nd Quartile 65 62 59 53 43 37 32 22 14 10 8 7 3 2 1 0
3rd Quartile 65 64 64 60 59 52 46 39 32 24 20 14 7 6 4 1
4th Quartile 65 65 64 62 61 59 56 51 40 37 29 23 18 15 7 2

$CAVG_{SG}$, sacituzumab govitecan average concentration; $CAVG_{tAB}$, total antibody average concentration; ORR, objective response rate; OS, overall survival; PFS, progression-free survival; pts, patients.
[a]Gray circles indicate data from individual pts. Closed squares (error bars) show observed proportion of pts. [b]Based on the primary PFS analysis and second OS interim analysis.
Dashed vertical lines in A indicate boundaries of the exposure quartiles. Dashed lines in C and D indicate median OS and PFS for each quartile.

Exposure-safety analysis

- The probability of any-grade events of all evaluated AEs and the risk of dose reductions or delays significantly increased with increasing $CAVG_{SG}$. Pts with *UGT1A1* genotype *28/*28 had a slightly higher risk of neutropenia **(Figure 2B)**
- Increase in $CAVG_{SG}$ was significantly associated with increased probability of grade ≥ 3 neutropenia and febrile neutropenia **(Table 4)**. *UGT1A1* *28/*28 genotype was associated with a greater risk of dose reductions and higher baseline body weight was associated with a reduced risk of dose delays (data not shown)

Table 4. Model-predicted odds ratio for AEs associated with an increase in SG exposure

AE	OR (95% CI) for increase in $CAVG_{SG}$ by 10%	AE (any grade)	OR (95% CI) for a 1 µg/mL increase in $CAVG_{SG}$
Neutropenia (any grade)	1.39 (1.33-1.45)	Diarrhea	1.40 (1.32-1.50)
Neutropenia (grade ≥ 3)	1.35 (1.30-1.41)	Nausea	1.37 (1.28-1.46)
Febrile neutropenia (grade ≥ 3)[a]	2.21 (1.86-2.64)	Vomiting	1.29 (1.22-1.37)
		Hypersensitivity	1.28 (1.21-1.35)

AE, adverse event; $CAVG_{SG}$, sacituzumab govitecan average concentration; CI, confidence interval; OR, odds ratio; SG, sacituzumab govitecan.
[a]All febrile neutropenia events were reported as grade 3 or 4 only.

This presentation is the intellectual property of the author/presenter. Contact them at abhishek.sathe@gilead.com for permission to reprint and/or distribute.